P67 IN VITRO CO-CULTURE MODEL SHOWS ADRENAL CELLS CAN DRIVE PROSTATE CANCER GROWTH IN ANDROGEN DEPRIVED CONDITIONS

J.M. Moll[1], W. Teubel[1], R. Graeser[2], I. Hickson[2], G. Jenster[1], W. M. van Weerden[1]

[1]Department of Urology, Erasmus MC, Rotterdam, Netherlands [2]Discovery and Development, Janssen Pharmaceuticals, Beerse, Belgium

Erasmus MC University Medical Center Rotterdam

Background & Aim

Prostate cancer (PC) growth is driven by testosterone (T) via the androgen receptor (AR). The initial treatment of metastatic PC is (chemical) castration. Despite castrate serum T levels, patients develop castration-resistant PC (CRPC) within 1-3 years. Studies indicate that CRPC is able to synthesise its own T. It is still debated, however, whether these T tumour levels originate from de novo synthesis or via conversion of circulating adrenal steroids. Abiraterone (Abi) is a new compound that blocks the CYP17A1 enzyme required for conversion of androgen precursors into adrenal androgens, and has shown increased survival of CRPC patients post-chemotherapy.

In this study, we used parental and CRPC cell lines to test if CRPC cells use de novo steroidogenesis or conversion of adrenal androgens to support AR-stimulated growth and created an in vitro co-culture model for adrenal stimulated cell growth.

Materials & Methods

- VCaP cells were used to model hormone-dependent PC. CRPC clones of VCaP were generated by long term culture in steroid-depleted medium with or without anti-androgens.
- Adrenal androgen producing H295R adrenal carcinoma cells were used to model adrenal androgen production.
- PC cells were incubated with androgen precursors, adrenal androgens or dihydrotestosterone(DHT) at supraphysiological levels or similar to those found in found in healthy human males. Cells were subsequently treated with Abi or anti-androgens.
- For co-culture experiments, H295R cells and PC cells were seeded separately using 24-wells plates with inserts.
- Cell growth was assessed by MTT assay.

Conclusion

Our results show that parental VCaP and VCaP CRPC cells do not depend on de novo steroidogenesis to survive in a testicular androgen-depleted environment. Androgens secreted by the adrenal cell line H295R induced growth in both parental and CRPC cells, which could be partially inhibited by Abi, validating previous reports that conversion of adrenal androgens is the main driver for CRPC.

Acknowledgements

The research leading to these results has received support from the Innovative Medicines Initiative Joint Undertaking under grant agreement n° 115188, resources of which are composed of financial contribution from the European Union's Seventh Framework Programme (FP7/2007-2013) and EFPIA companies in kind contribution. Development of CRPC clones was supported by TI-Pharma Project T3-107

Results

CYP17A1 is a critical for androgen production

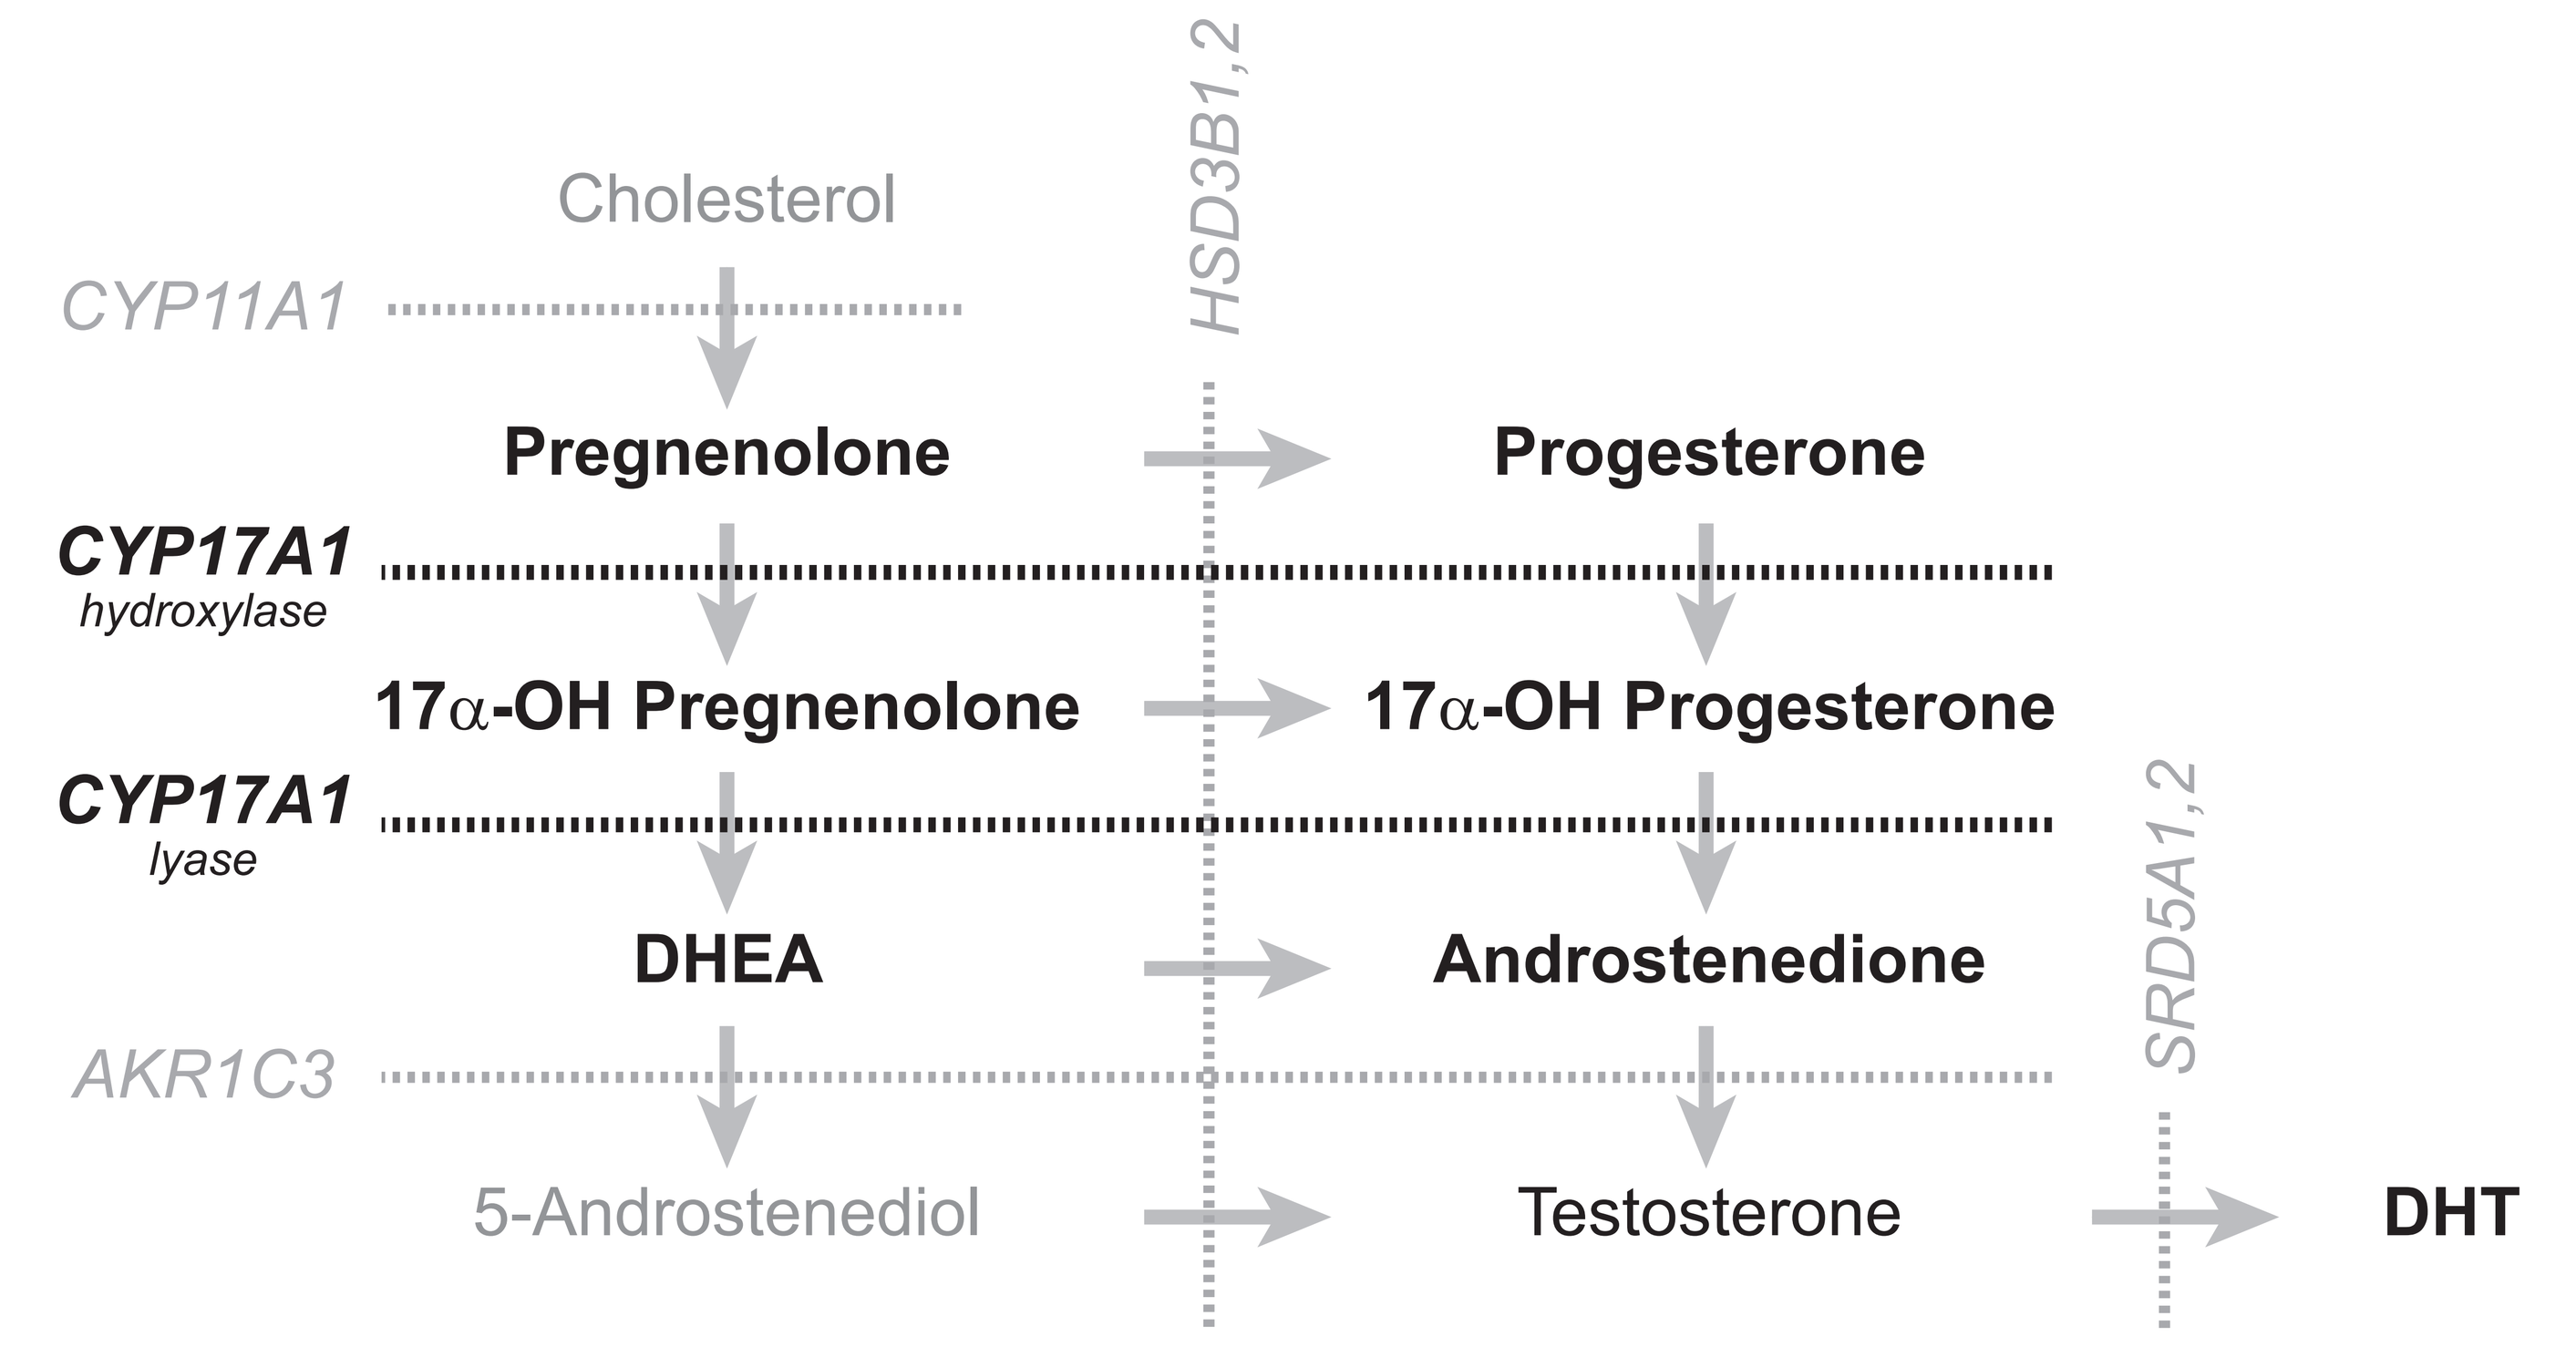
Cholesterol
CYP11A1
HSD3B1,2
Pregnenolone
Progesterone
CYP17A1
hydroxylase
17α-OH Pregnenolone
17α-OH Progesterone
CYP17A1
lyase
DHEA
Androstenedione
SRD5A1,2
AKR1C3
5-Androstenediol
Testosterone
DHT

▲ **Figure 1: Overview of the steroidogenic pathway in humans.** After cholesterol is converted into pregnenolone, CYP17A1 hydroxylates and lyates it to 17OH-pregnenolone and DHEA. HSD3-enzymes can metabolize these precursors into progesterone, 17OH-progesterone and androstenedione, respectively. DHEA and androstenedione can subsequently be converted into testosterone and DHT. In humans, CYP17A1 actvity is present in the testicular and adrenal glands. In mice CYP17A1 is only expressed in testicular tissue.

Androgen precursors induce PC growth *in vitro* regardless of CYP17A1 activity

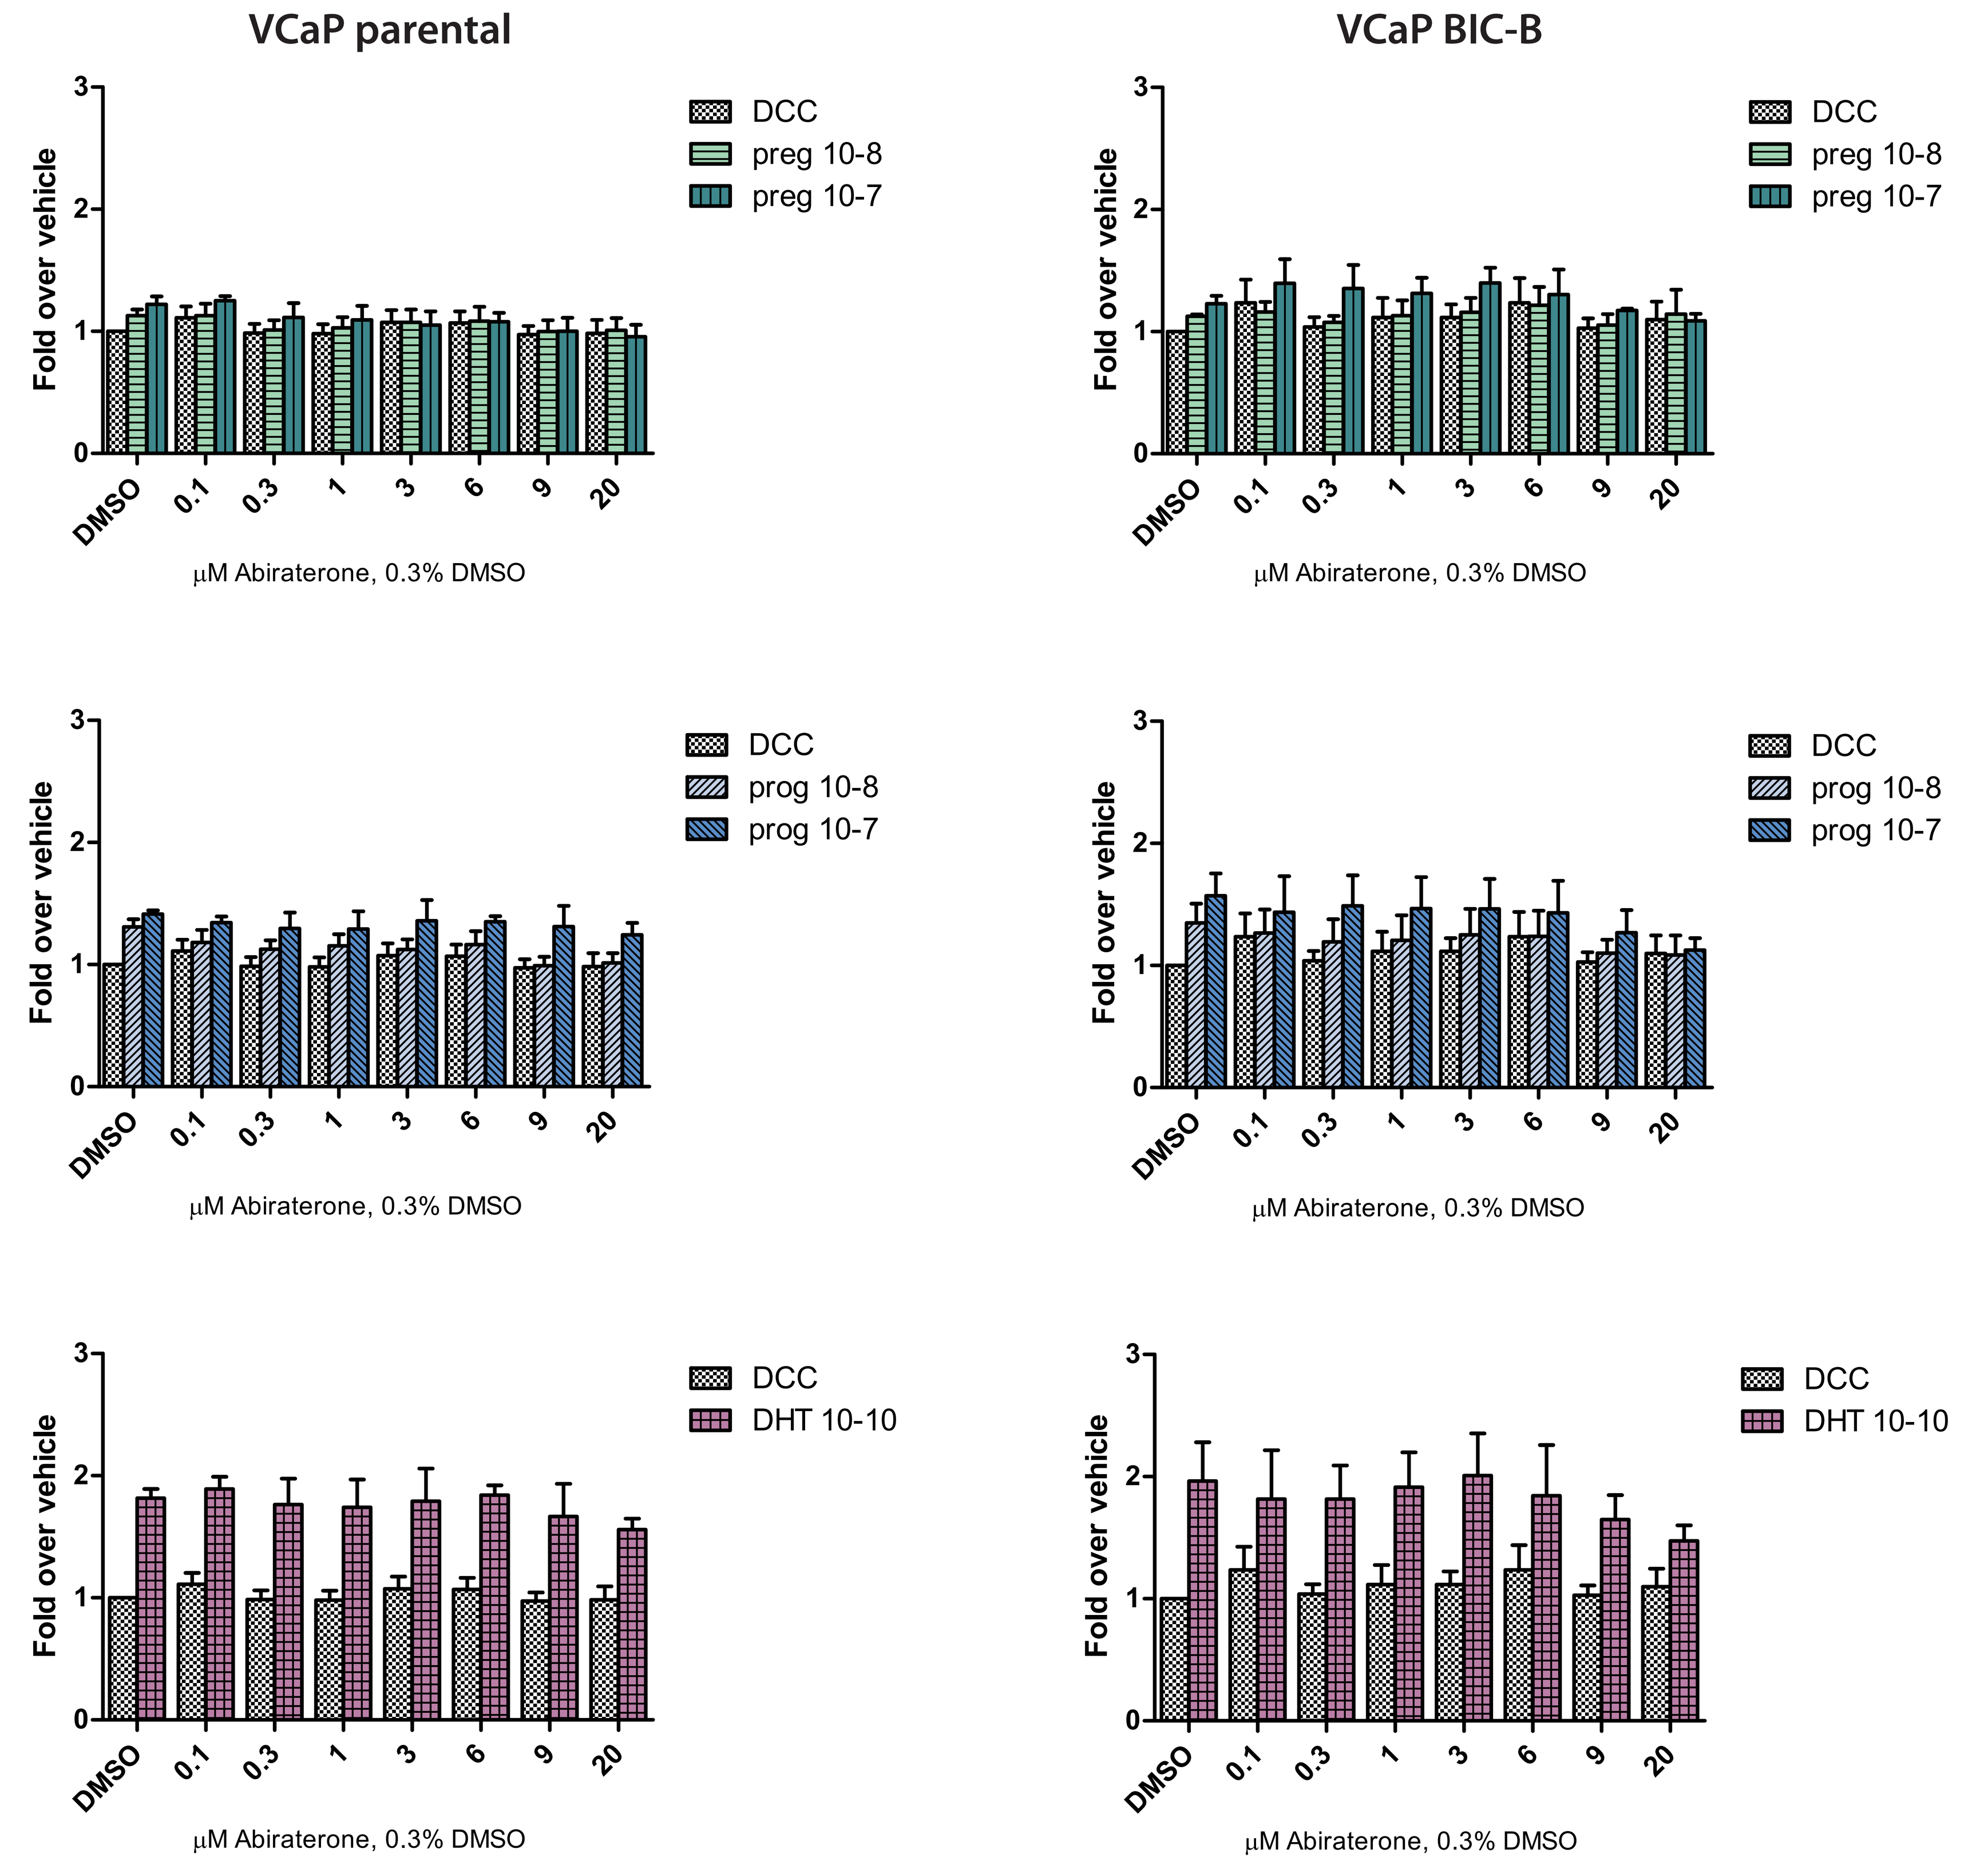
VCaP parental
VCaP BIC-B
Fold over vehicle
DCC
preg 10-8
preg 10-7
prog 10-8
prog 10-7
DHT 10-10
DMSO 0.1 0.3 1 3 6 9 20
μM Abiraterone, 0.3% DMSO

▲ **Figure 2: Androgen precursors induce minor growth in castration-naïve and CRPC cells. Left:** MTT-results in VCaP parental cells. Top: pregnenolone at 100 or 10 nM induce little growth, which is inhibited at higher concentrations of Abi. Middle: progesterone is more potent at inducing growth, wich is nog inhibited at concentrations of Abi that adequately block CYP17-activity (0.1 μM). Bottom: DHT (0.1 nM) induced growth is inhibited at higfher levels of Abi, suggesting a direct AR-blocking property of Abi. **Right:** like parental cells, VCaP FLU-D cells (cultered in steroid stripped medium and 1 μM of the anti-androgen OH-Flutamide) grow more in the presence of pregnenolone or progesterone, despite adequate CYP17A1 blockade. At higher levels of Abi, DHT-induced growth was inhibited.

Physiological levels of adrenal androgens, but not androgen precursors drive castration-naïve and CRPC cell growth

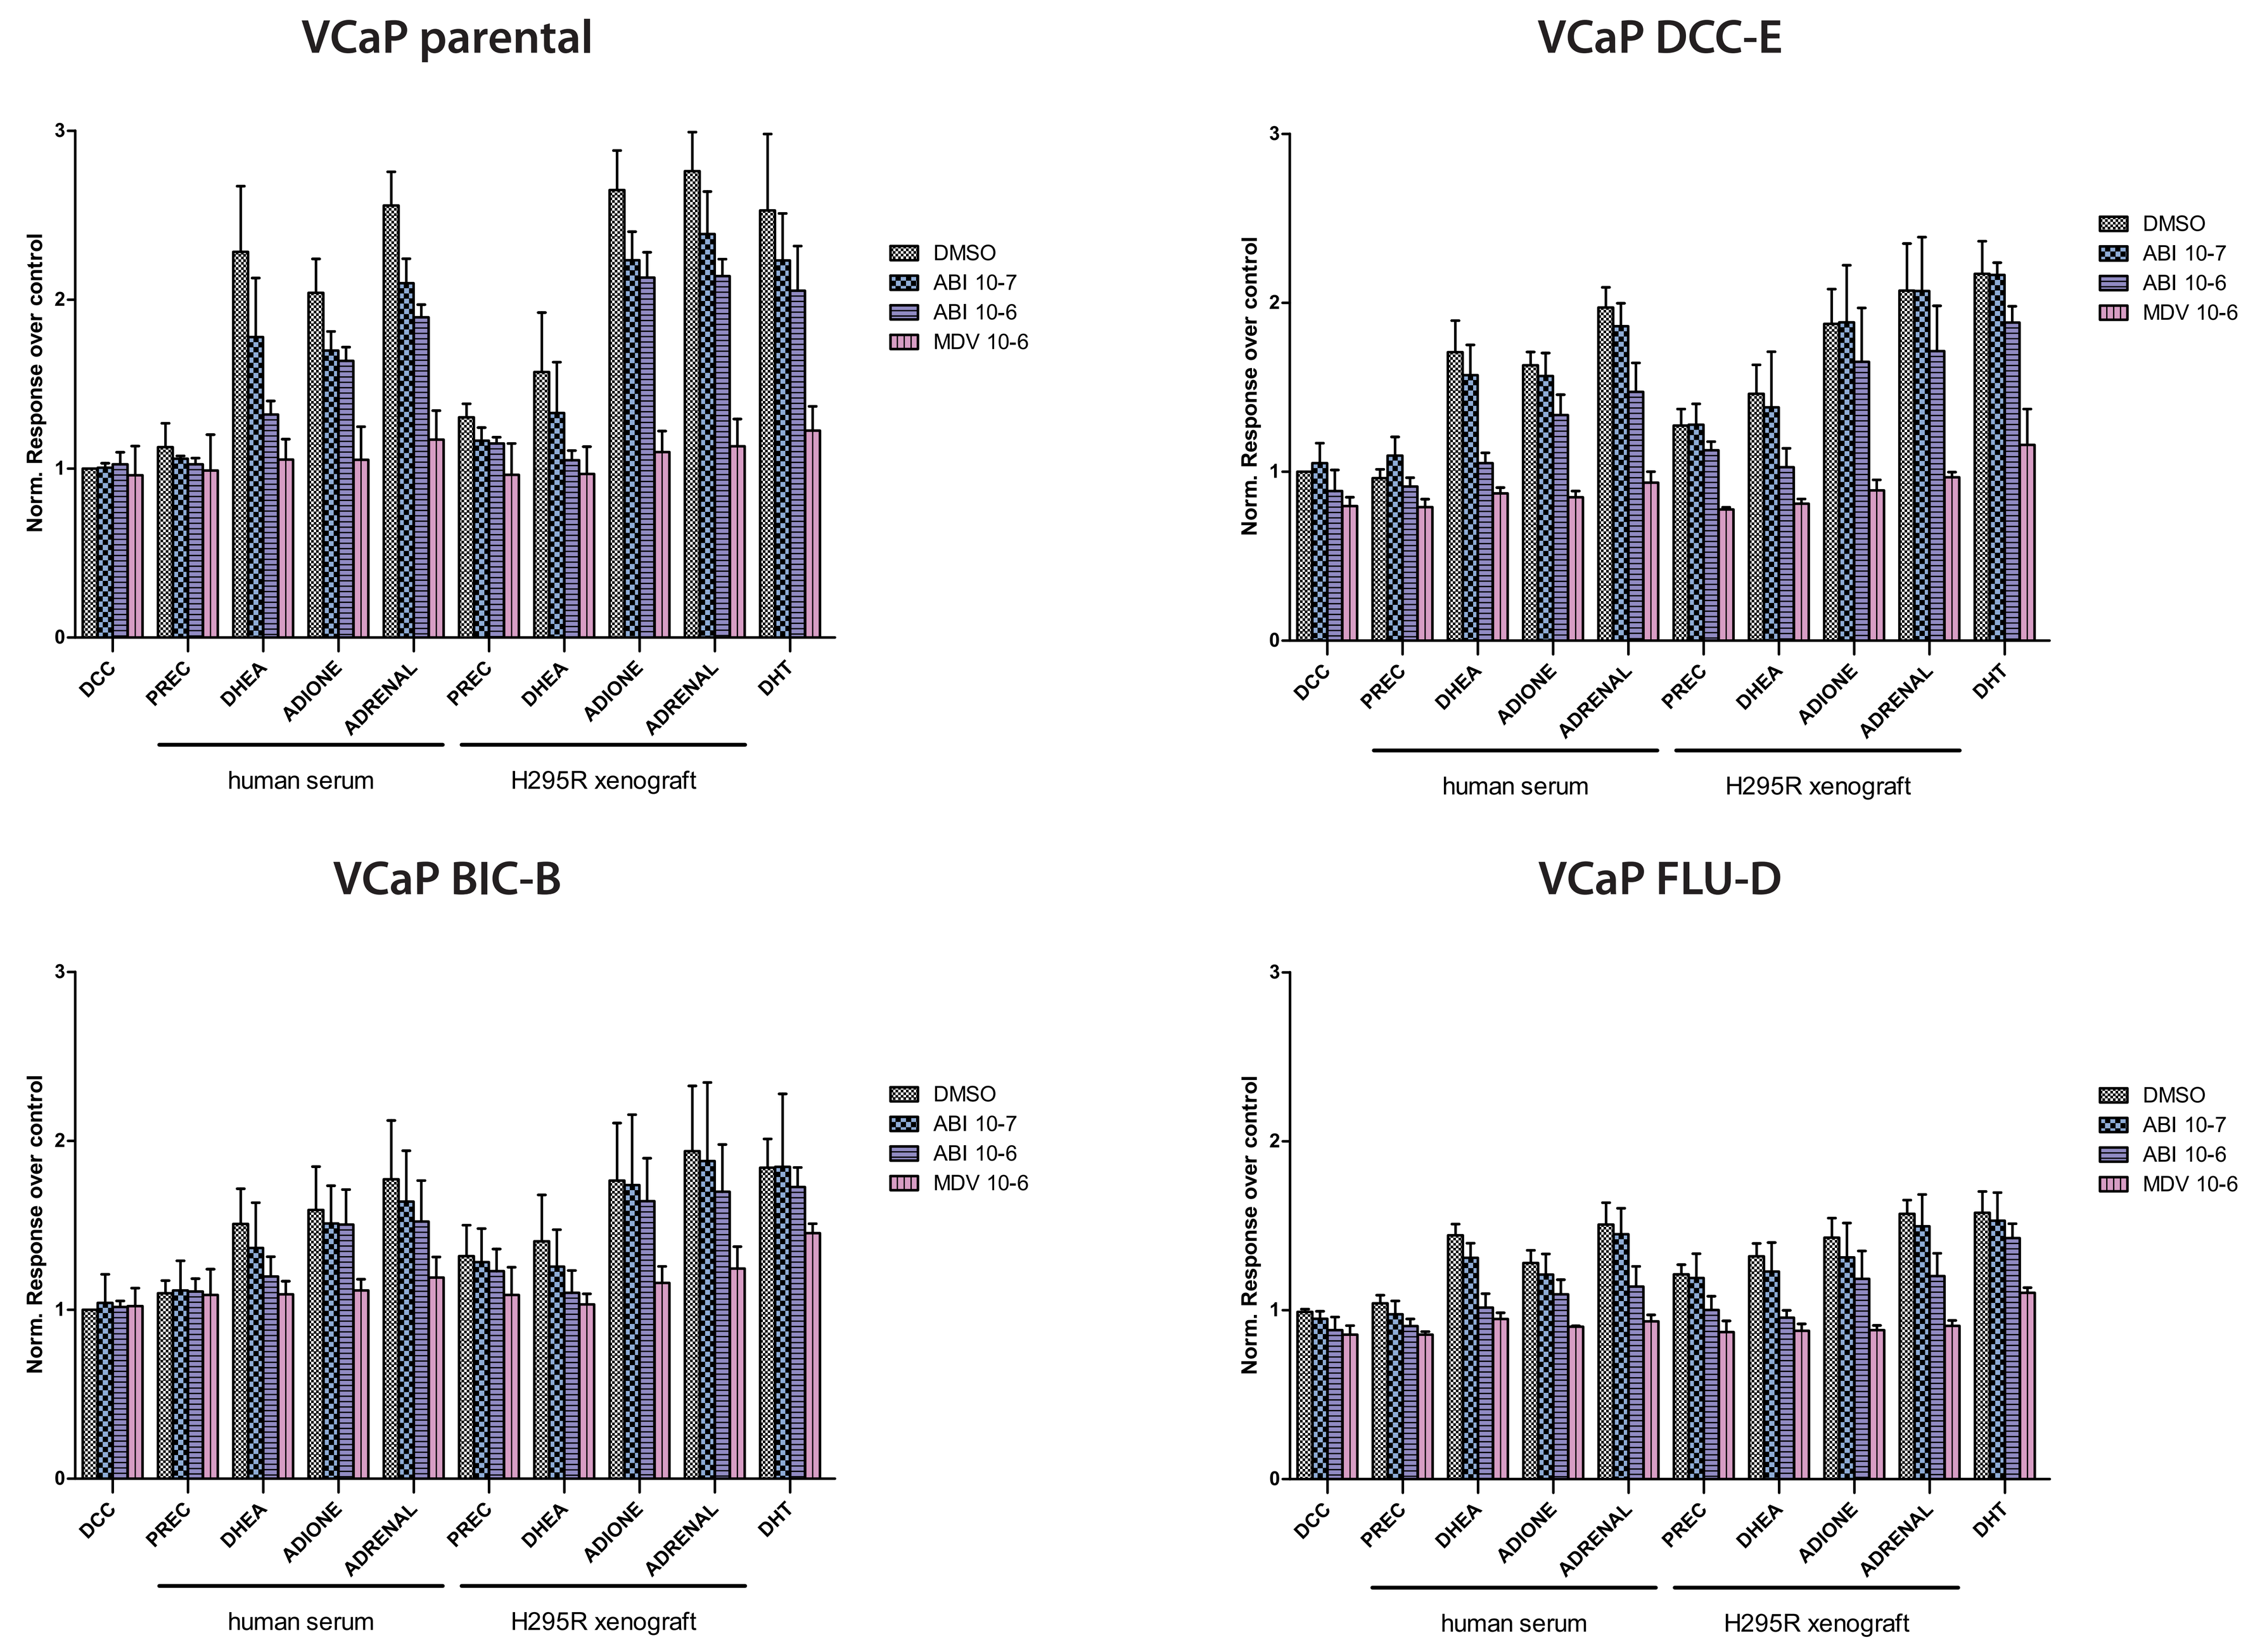
VCaP parental
VCaP DCC-E
VCaP BIC-B
VCaP FLU-D
Norm. Response over control
DMSO
ABI 10-7
ABI 10-6
MDV 10-6
DCC PREC DHEA ADIONE ADRENAL PREC DHEA ADIONE ADRENAL DHT
human serum
H295R xenograft

▲ **Figure 3: Castration-naïve and castration-resistant PC cells react similarly to levels of steroids found in the average 60 year-old male and to those found in H295R xenograft-bearing mice.** In VCaP parental and VCaP-derived CRPC cell lines, no significant growth induction was observed in the presence of preg and prog. levels of adrenal androgens induced growth which could be inhibited by the novel anti-androgen MDV310, but to a lesser extent by Abiraterone. *Steroid levels in nM (60 y.o. male/H295R xenograft): pregnenolone 1.5/30 ; progesterone 0.5/5 ; DHEA 10/5 ; androstenedione 2.5/7.5 ; DHT 0.1.*

Co-culture with H295R adrenal carcinoma cells can drive PC growth in castrate conditions

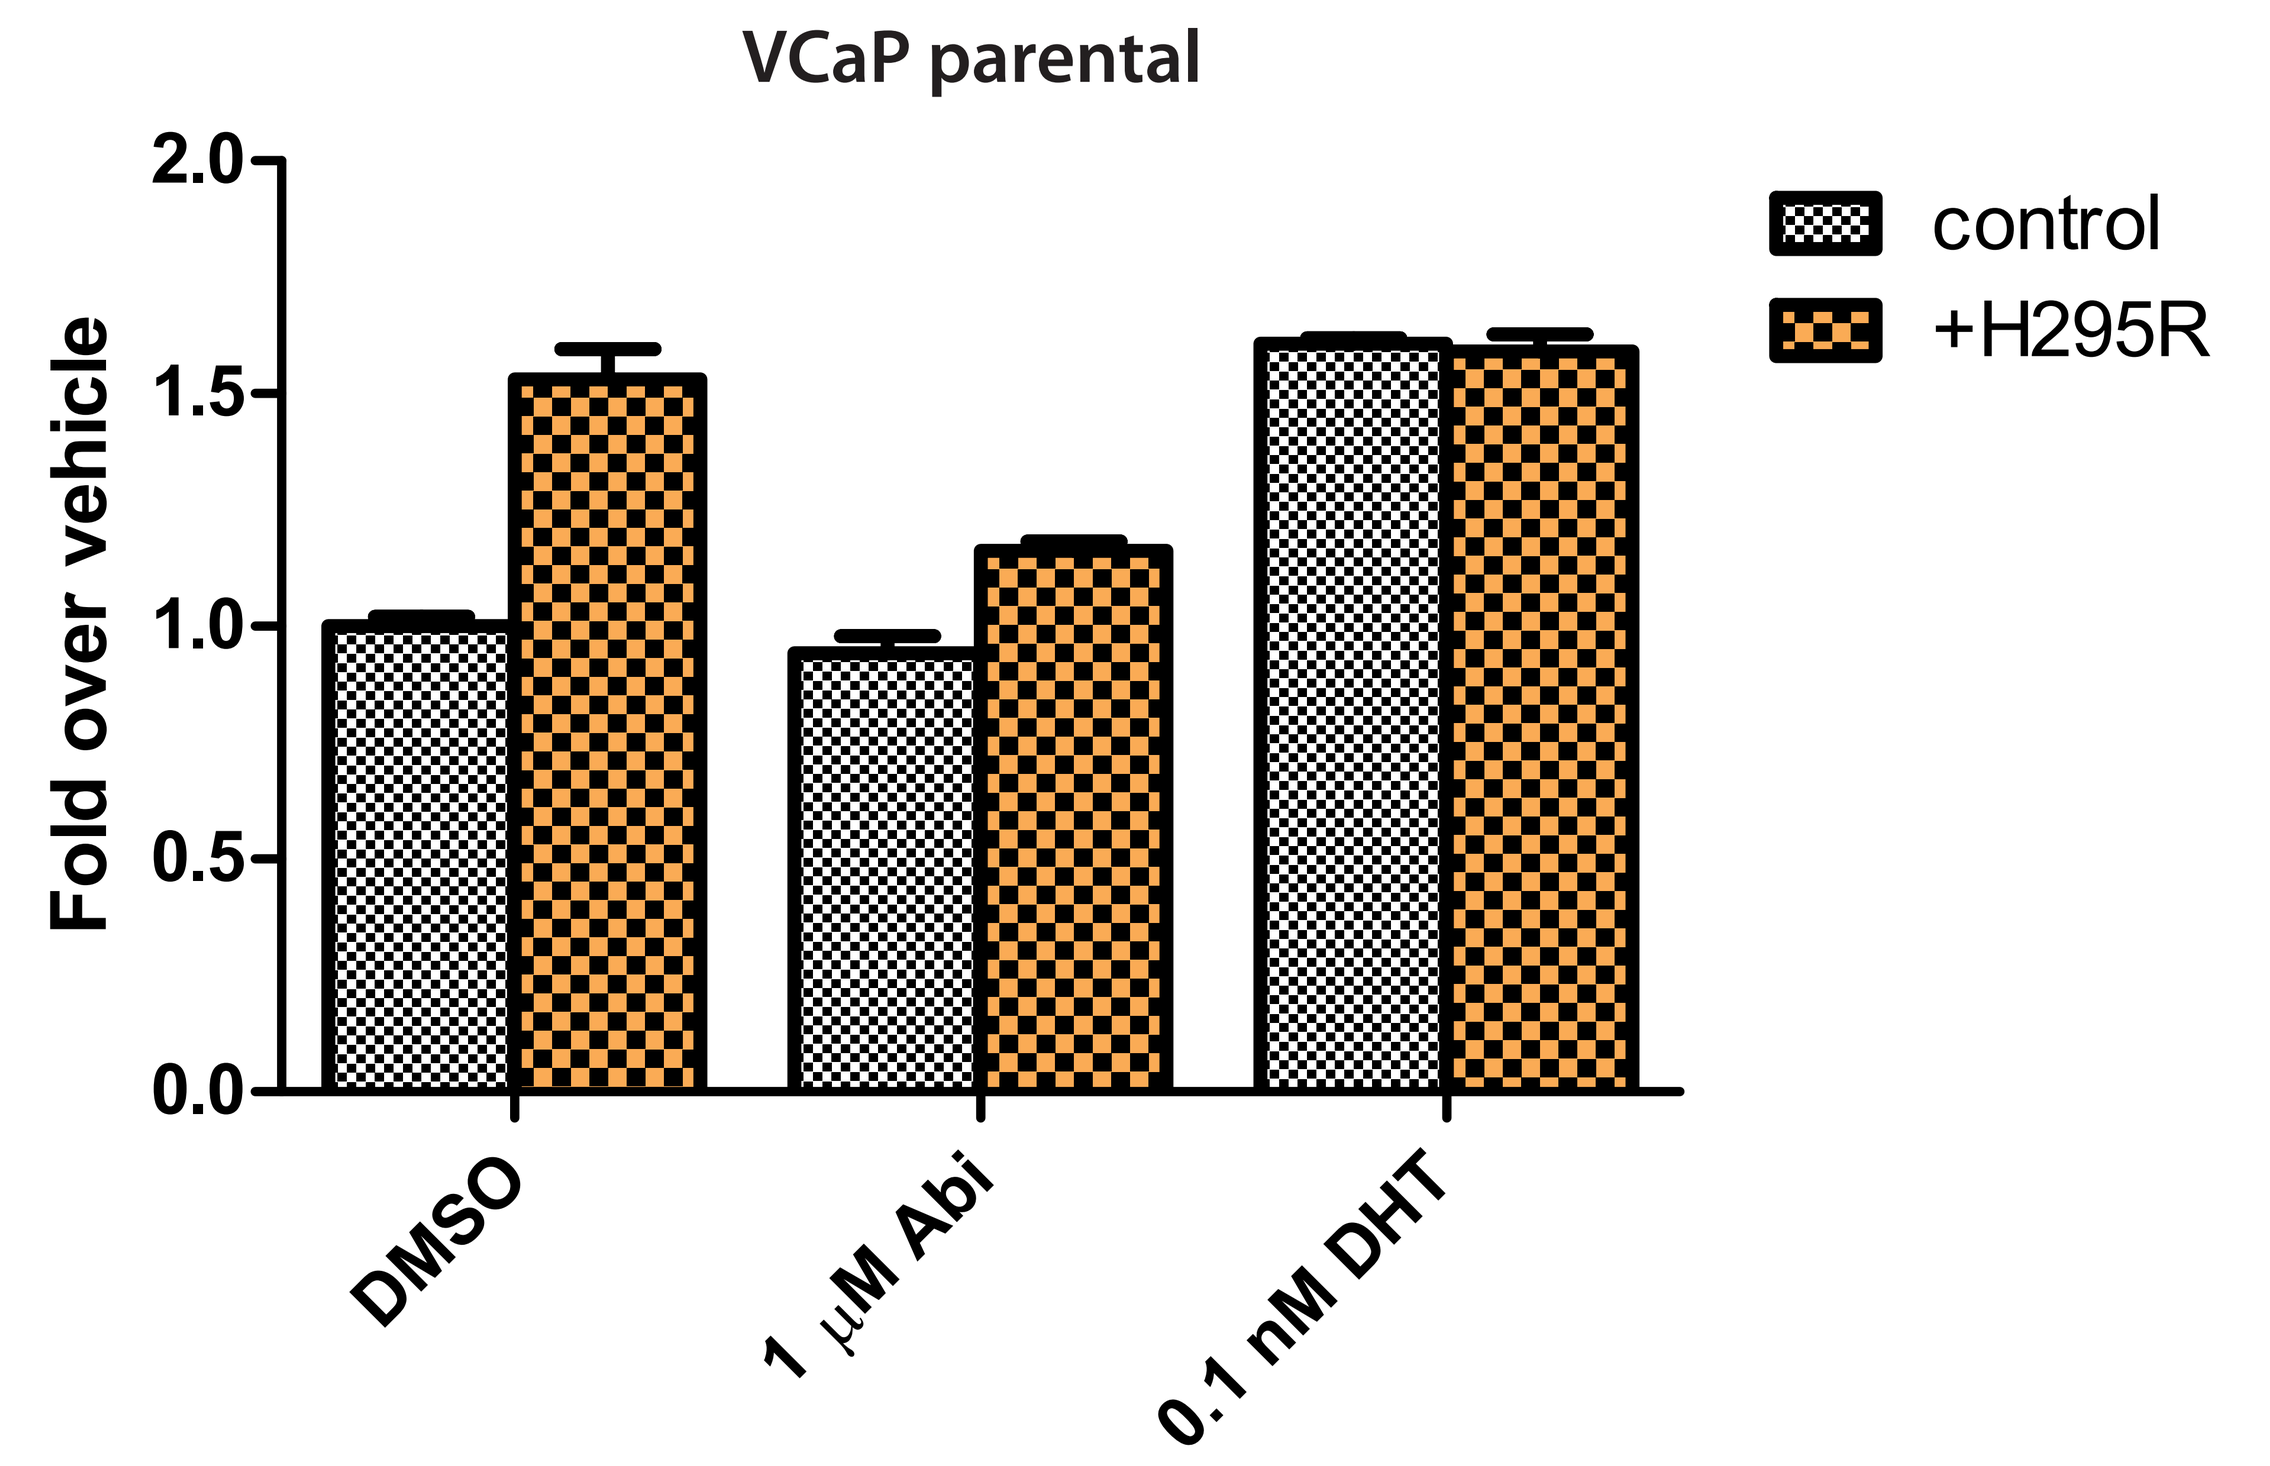
VCaP parental
Fold over vehicle
control
+H295R
DMSO
1 μM Abi
0.1 nM DHT

◄ **Figure 4: H295R adrenal cells stimulate VCaP cell growth in castrate conditions.** Co-culture with adrenal cells stimulated PC cell growth. 1 uM Abi could partially inhibit this growth stimulus, suggesting that steroid synthesis in the adrenals, rather than *de novo* synthesis in PC cells is more important for AR-activation in CRPC.

Future plans

To model CPRC *in vivo* we are planning to move forward to study tumour growth in a murine xenograft model. To replicate the hormonal environment that is normally maintained by human adrenals, H295R xenografts will be transplanted simultaneously to secrete the normal adrenal hormones found in men, but not in mice.